A NOVEL DUAL NLRP1 AND NLRP3 INFLAMMASOME INHIBITOR FOR THE TREATMENT OF INFLAMMATORY DISEASES

CENTRE FOR INNATE IMMUNITY AND INFECTIOUS DISEASES HUDSON INSTITUTE OF MEDICAL RESEARCH

Callum AH Docherty[1,2], Anu A Fernando[3], Sarah Rosli[1,2], Maggie Lam[1,2], Michelle D Tate[1,2], Chris Murphy[4], Adriano G Rossi[3], Ashley Mansell[1,2,4]

1 Centre for Innate Immunity and Infectious Diseases, Hudson Institute of Medical Research, Clayton, VIC, Australia, 2 Department of Molecular and Translational Sciences, Monash University, Clayton, VIC, Australia, 3 University of Edinburgh Centre for Inflammation, Queen's Medical Research Institute, Research, Edinburgh BioQuarter, Edinburgh, United Kingdom, 4 Adiso Therapeutics, 530 Virginia Road, Suite 300, Concord, MA 01742, USA

INTRODUCTION

- Excessive inflammasome activation is associated with the pathophysiology of a broad range of inflammatory diseases. NLRP3-induced inflammation is associated with many diseases, notably neurodegenerative (Alzheimer's, Parkinson's), cardiovascular, metabolic (diabetes, obesity), pulmonary (chronic obstructive pulmonary disease (COPD), asthma) diseases and cancer.
- Significantly, it is becoming increasingly evident that NLRP1 dysfunction is also associated with skin-related genetic disorders, and similar to NLRP3, NLRP1 has increasingly been linked with neurodegenerative, pulmonary, metabolic, skin inflammatory and intestinal disease pathobiology.
- Given this prominent role in diseases by both NLRP1 and NLRP3, there is considerable interest in targeting inflammasome-mediated inflammation.
- To date, pharmacological targeting of inflammasomes has focused primarily upon NLRP3 activity, however, ***there is no specific inhibitor described for NLRP1***.
- Given the overlapping and complementary roles of both NLRP3 and NLRP1 in a wide variety of diseases, we sought to develop therapeutic targets to antagonize inflammasome activity as potential therapeutics to reduce inflammatory disease.

ADS032 INHIBITS BOTH NLRP1 and NLRP3

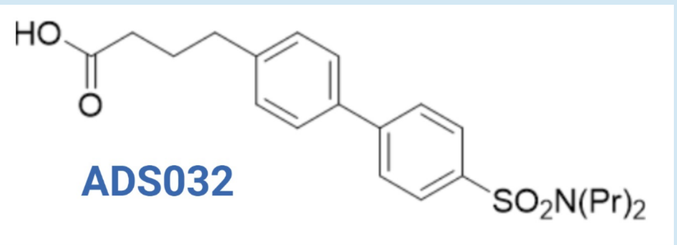
HO
O
ADS032
$SO_2N(Pr)_2$

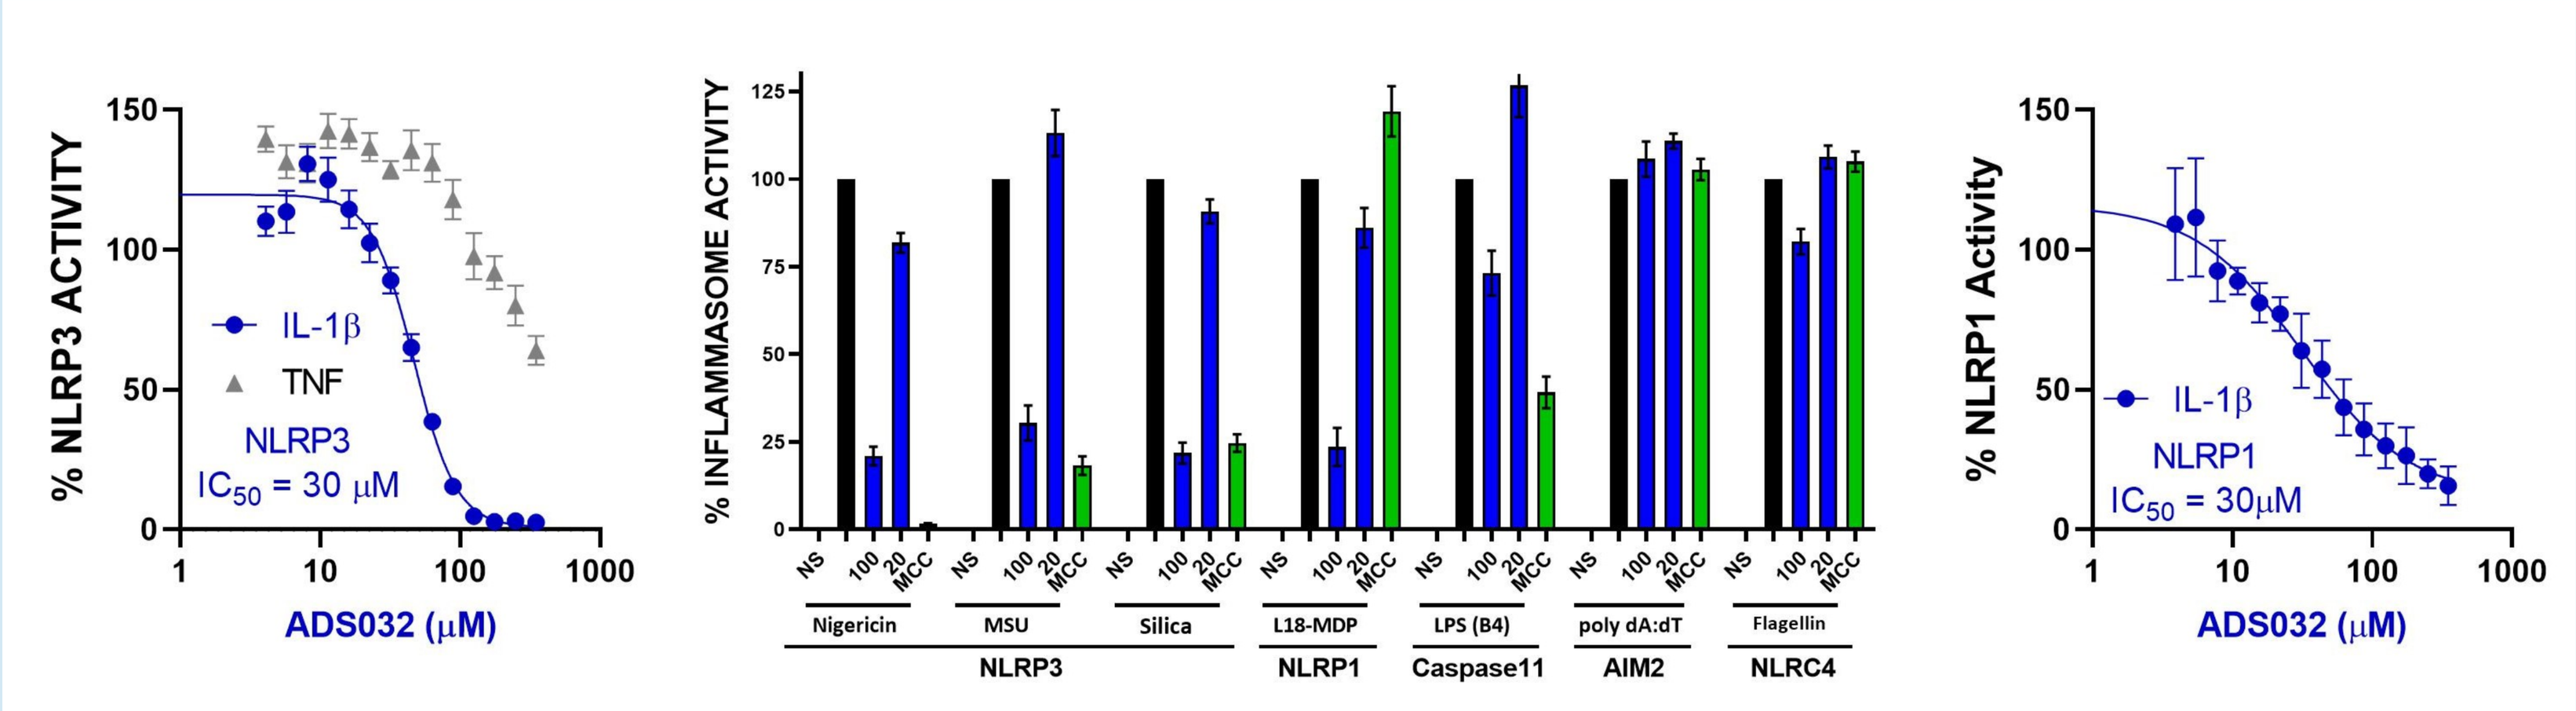
% NLRP3 ACTIVITY
IL-1β
TNF
NLRP3
IC_{50} = 30 μM
ADS032 (μM)
% INFLAMMASOME ACTIVITY
Nigericin
MSU
Silica
L18-MDP
LPS (B4)
poly dA:dT
Flagellin
NLRP3
NLRP1
Caspase11
AIM2
NLRC4
% NLRP1 Activity
IL-1β
NLRP1
IC_{50} = 30μM
ADS032 (μM)

ADS032 is the first in class dual NLRP3 and NLRP1 Inhibitor.

iBMDMs were primed with 100ng/ml LPS for 3 h, pre-treated with ADS032 (3.9-350 μM) for 60 mins prior to challenge with NLRP3 agonist Nigericin (3 μM) for 120 mins, or NLRP1 agonist L18-MDP (100 μg/ml) for 16 h. To determine ADS032 NLR specificity, primed iBMDMs were treated with **ADS032 (20, 100 μM),** or **MCC950 (5 μM)** where indicated for 60 mins prior to challenge with inflammasome agonists; **NLRP3**: nigericin , monosodium urate crystals; MSU, silica, **NLRP1**: L18-MDP, **Non-canonical**: transfected LPS Serotype 0111:B4, **AIM2**: poly dA:dT, or **NLRC4**: Flagellin. Data are mean ± SEM of three independent experiments carried out in triplicate (ie. n=9).

ADS032 TARGETS NLRP1 and NLRP3 PULMONARY INFLAMMATION

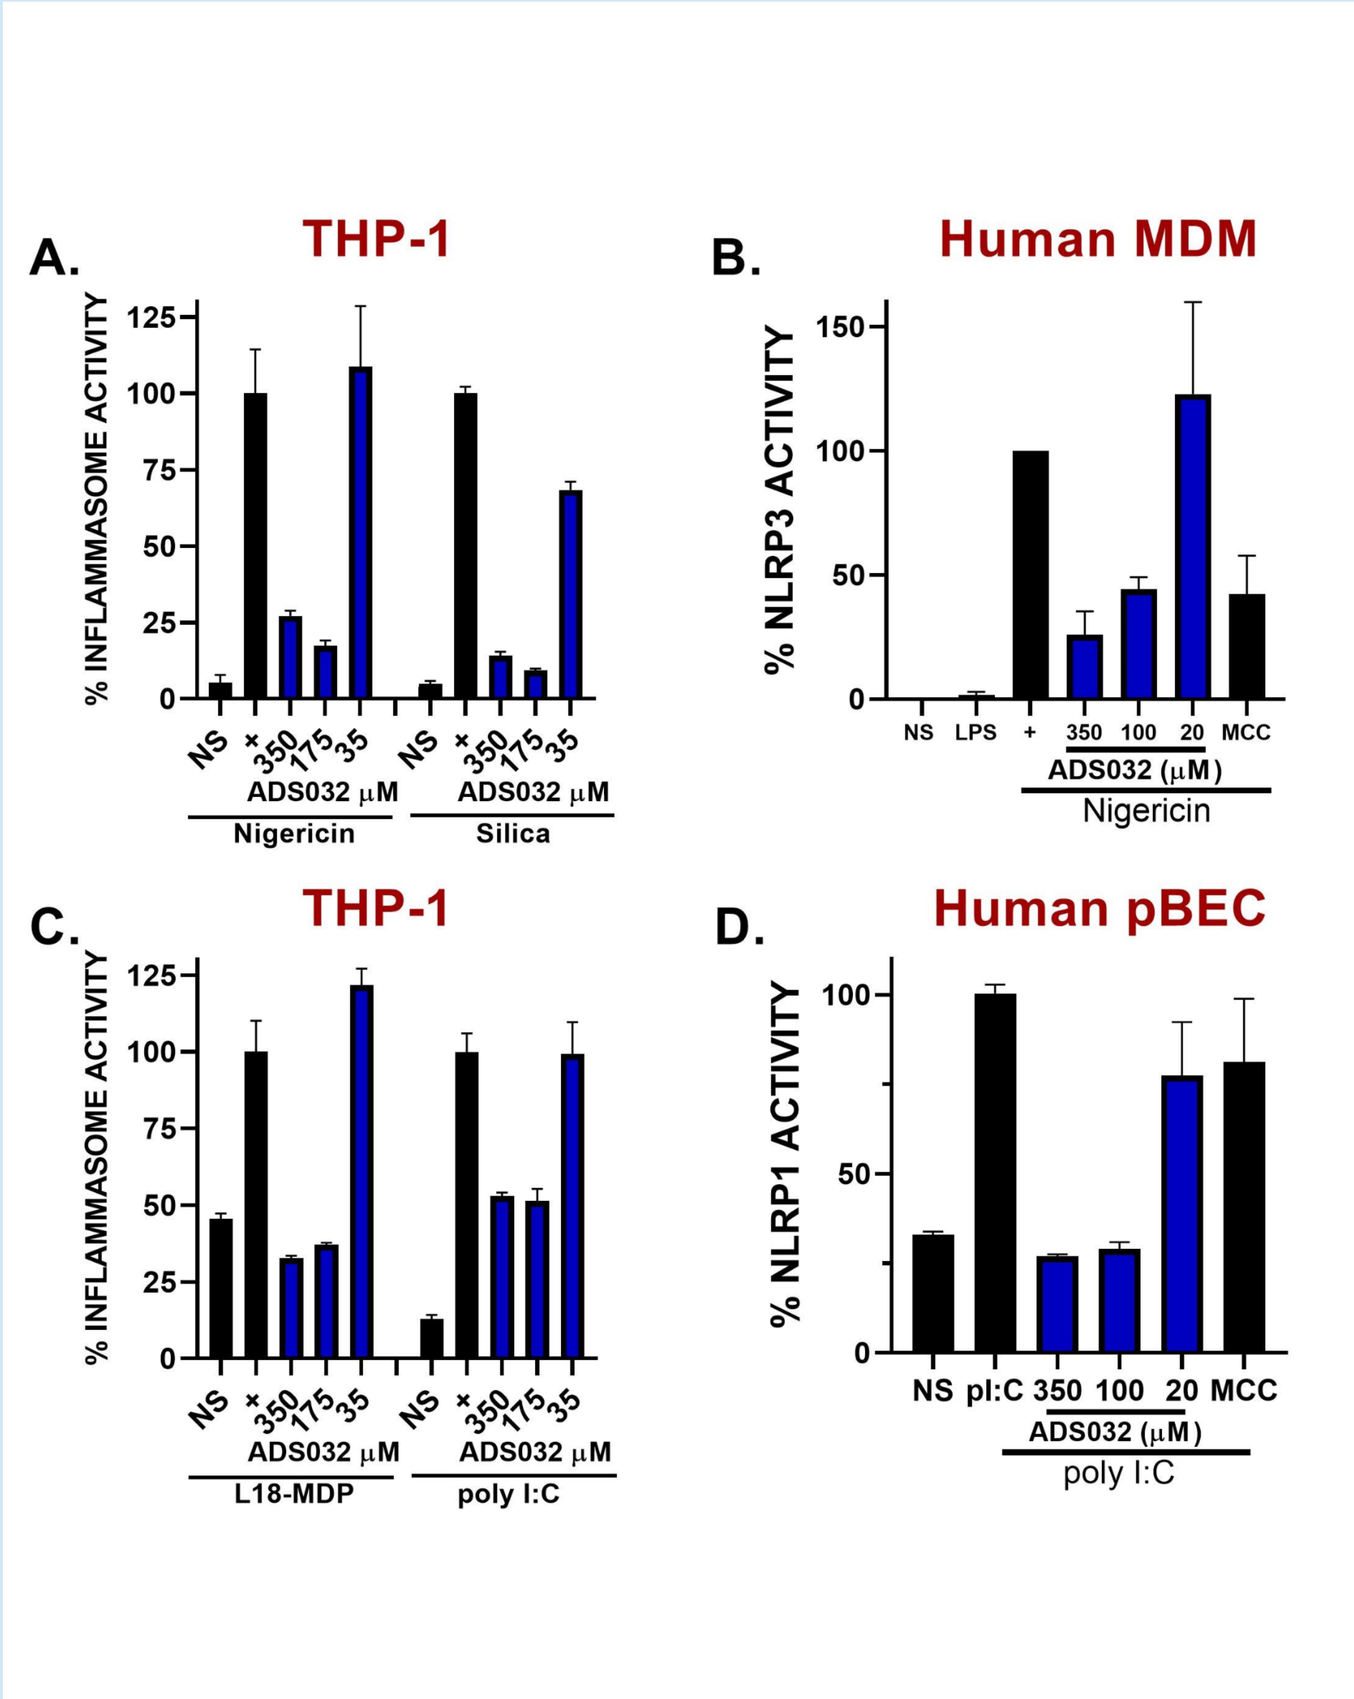
A. THP-1
B. Human MDM
C. THP-1
D. Human pBEC
% INFLAMMASOME ACTIVITY
% NLRP3 ACTIVITY
% NLRP1 ACTIVITY
ADS032 μM
Nigericin
Silica
L18-MDP
poly I:C

ADS032 effectively inhibits NLRP1 and NLRP3 inflammasome activity in human macrophages and bronchial epithelial cells.

PMA-differentiated THP-1 cells primed with LPS (100 ng/ml) for 3 h, pretreated or not with ADS032 (35-350 μM) for 60 min and then challenged with (**A**) NLRP3 agonists Nigericin (6 μM) or silica (250 μg/ml) for 120 min or 360 min; or (**C**) NLPR1 agonists L18-MDP (100 μg/ml) or transfected poly I:C (200 ng/ml) for 16 h or 8 h respectively.
(**B**) Human monocyte-derived macrophages were treated for 3 h with 50 pg/ml LPS, treated with ADS032 (20-350 μM) or **MCC950 (MCC; 5 μM)** for 60 min, then challenged with Nigericin (6 μM) for a further 120 min.
(**D**) Bronchial epithelial cells obtained from normal patients were treated with ADS032 as indicated for 60 min and treated with poly I:C for a further 8 h.

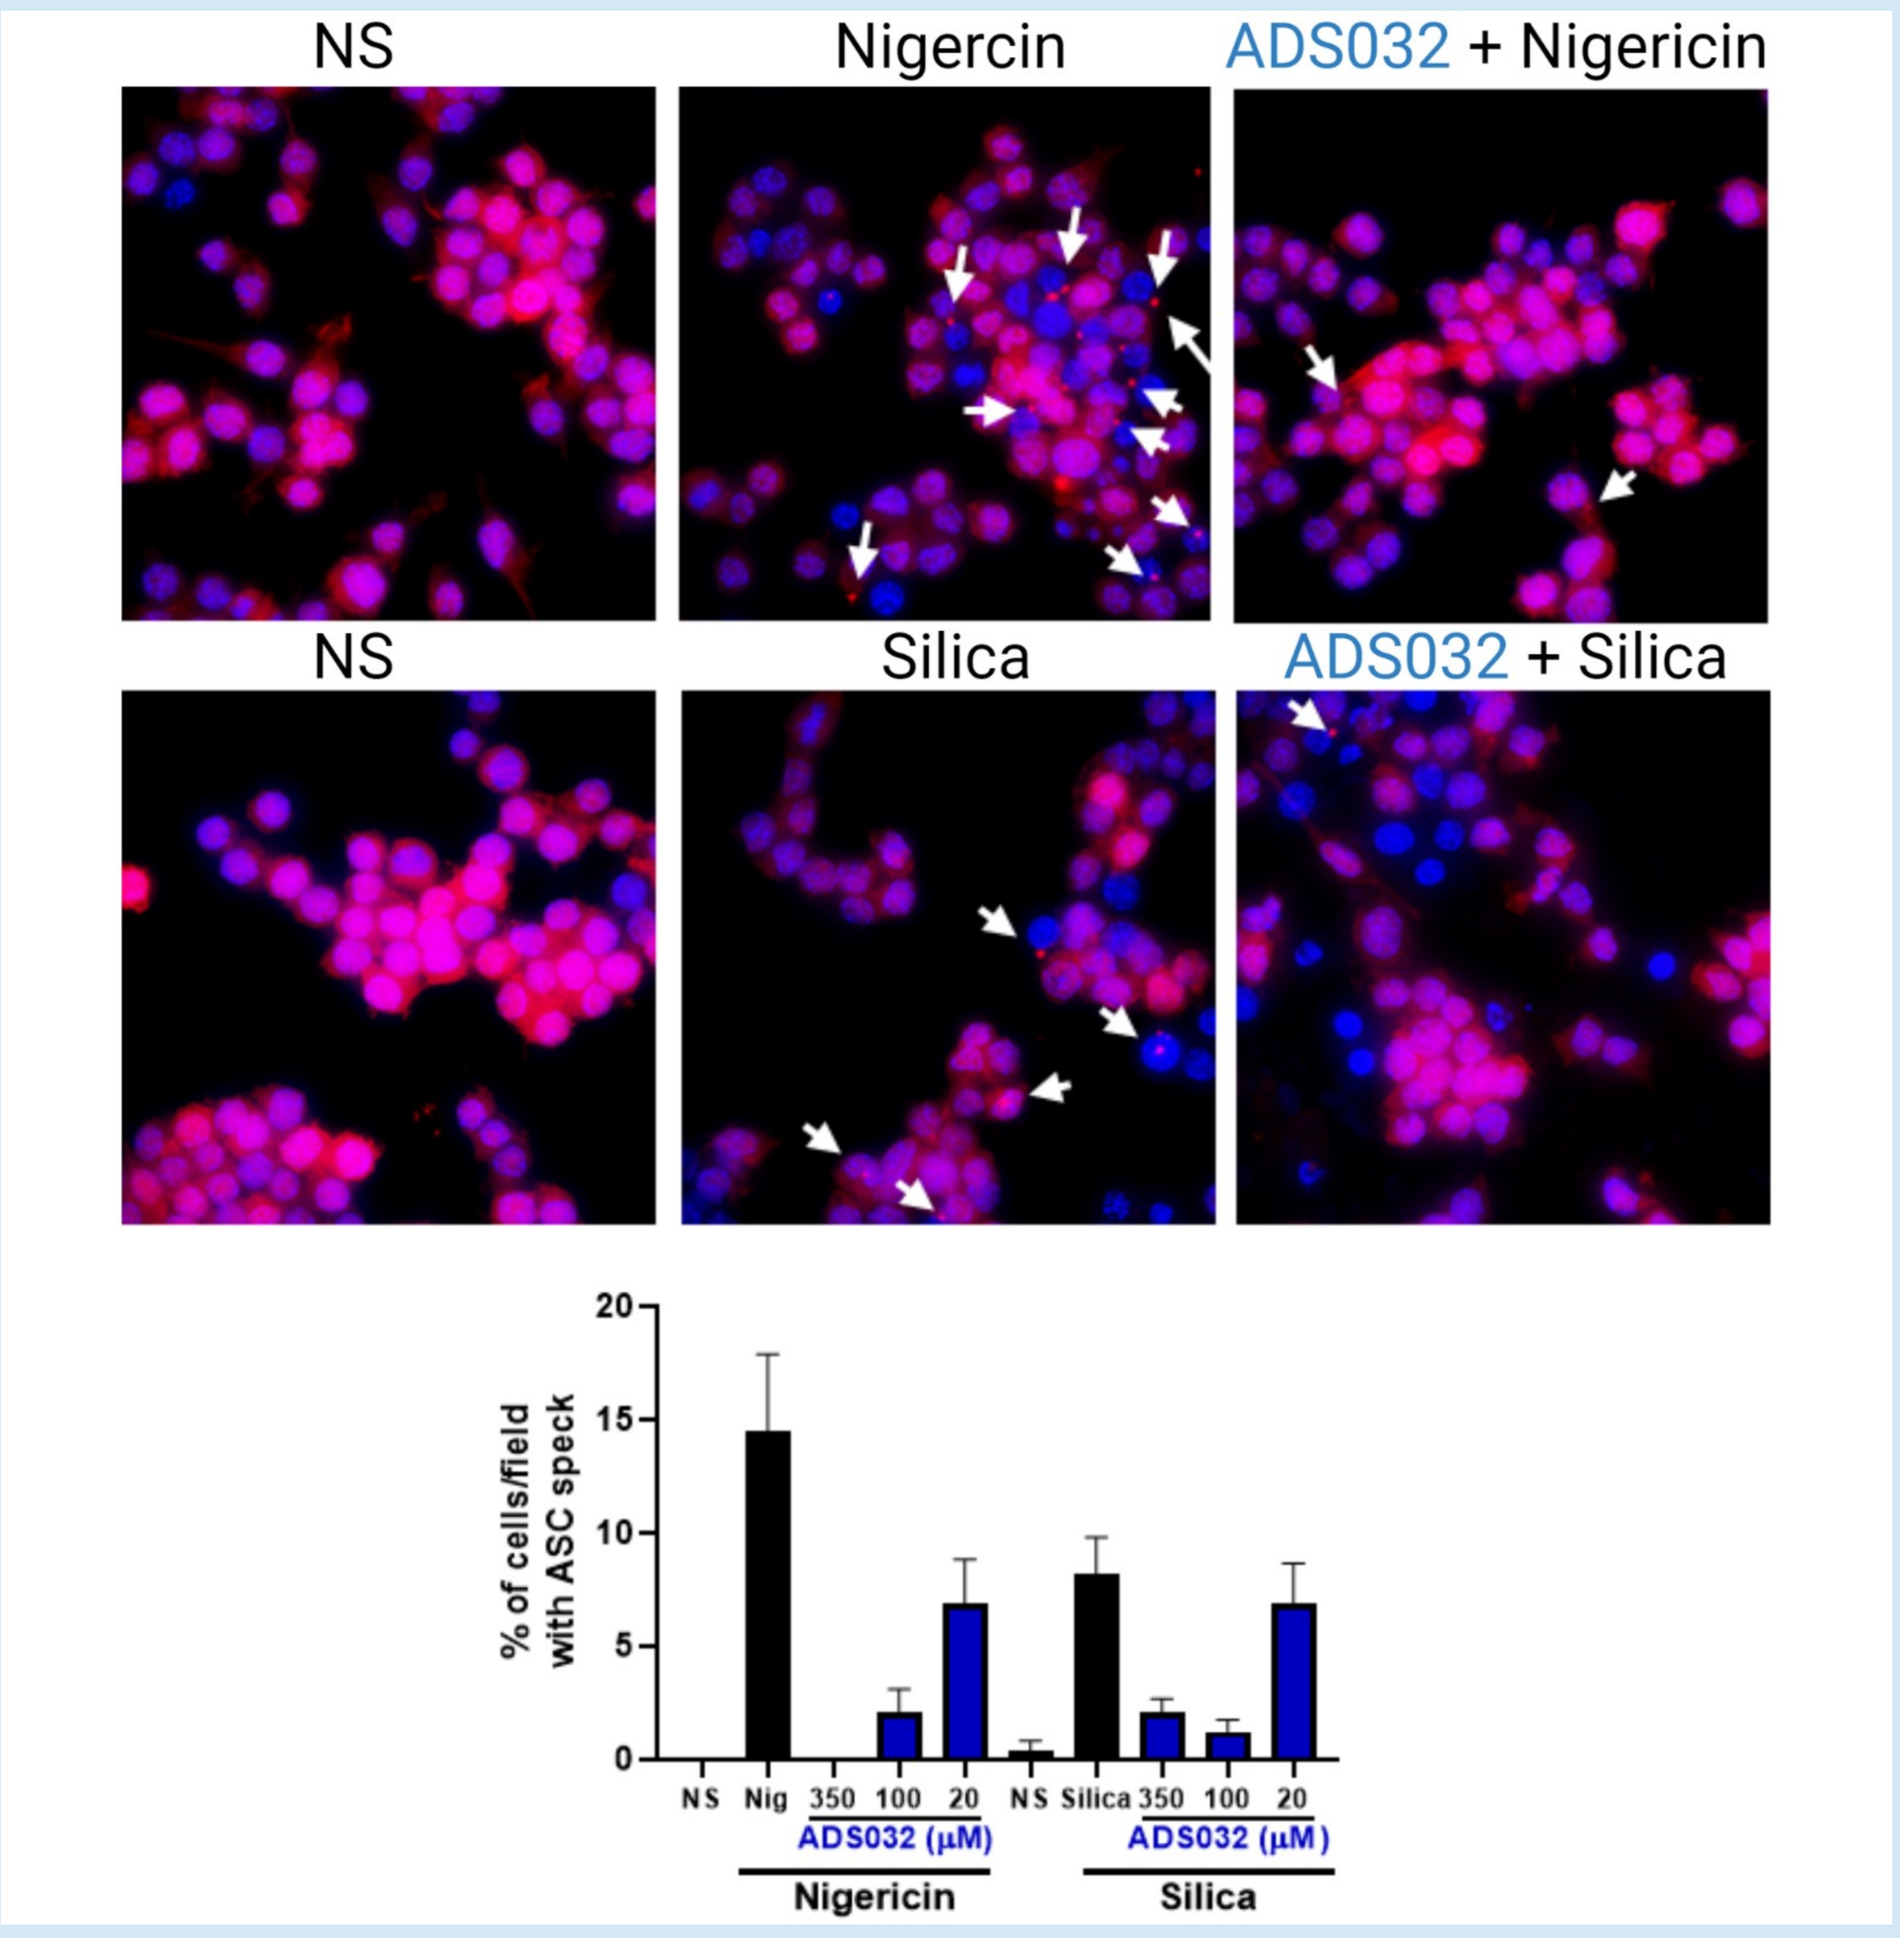
NS
Nigercin
ADS032 + Nigericin
NS
Silica
ADS032 + Silica
% of cells/field with ASC speck
ADS032 (μM)
Nigericin
Silica

ADS032 directly targets and inhibits formation of the NLRP3 oligomeric inflammasome complex.

ASC-tagged Cerulean (pseudo-colored **Red**) cells were treated or not with ADS032 (20, 100 and 350 μM) for 60 min and then challenged with either Nigericin (6 μM, 120 min) or silica (250 μg/ml, 5 h) before fixation. Cells were stained with **DAPI (Blue)** to identify cell nuclei. Images are flattened maximum-intensity projections of 3-dimensional deconvolved *z* stacks. Specks (white arrows) were observed with confocal microscopy the percentage of specks per field of view (9 fields per treatment group) were counted and compared to agonist-treated cells.

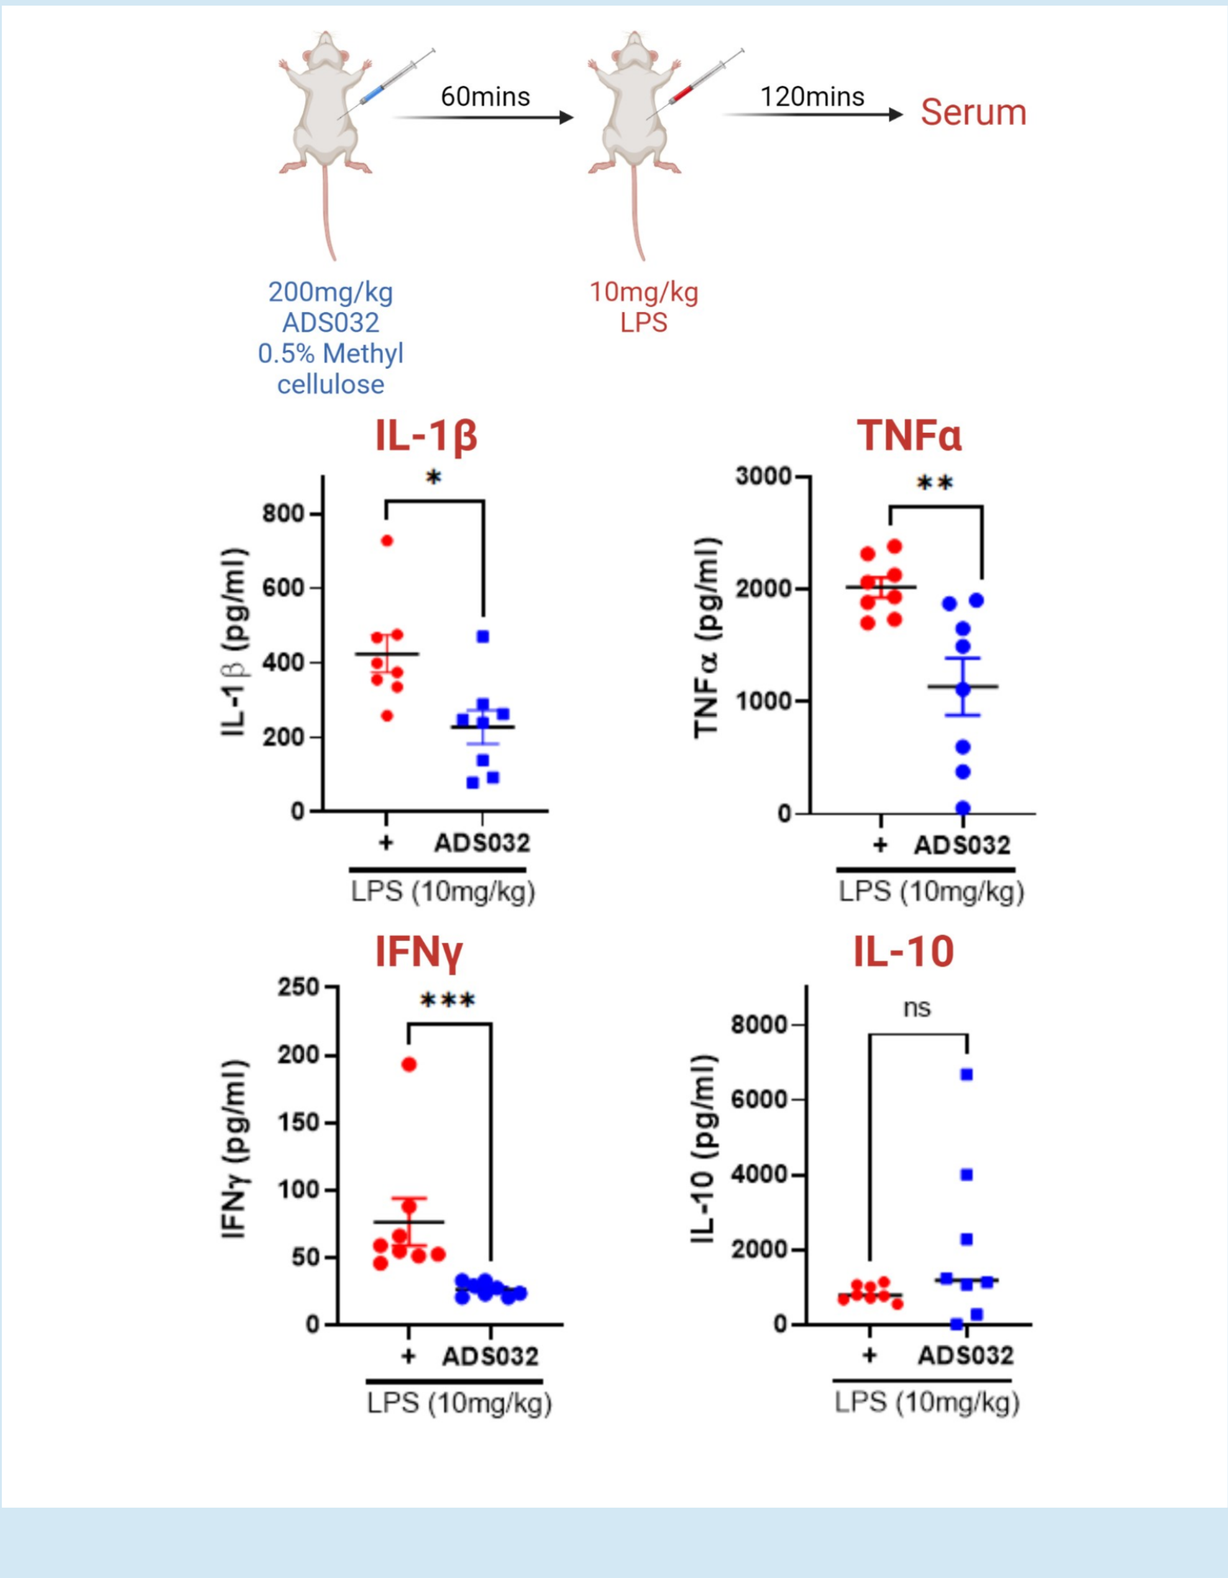
60mins
120mins
Serum
200mg/kg ADS032 0.5% Methyl cellulose
10mg/kg LPS
IL-1β
TNFα
IFNγ
IL-10
LPS (10mg/kg)

ADS032 reduces LPS-induced inflammation in vivo.

C57Bl/6 WT n=8 mice/grp (4M/F) were i.p. pretreated with ADS032 (200 mg/kg) or vehicle control (methylcellulose) for 60 min prior to LPS challenge (10 mg/kg i.p.) for 120 min. Student's t-test, *p < 0.05, **p < 0.01, ***p < 0.001

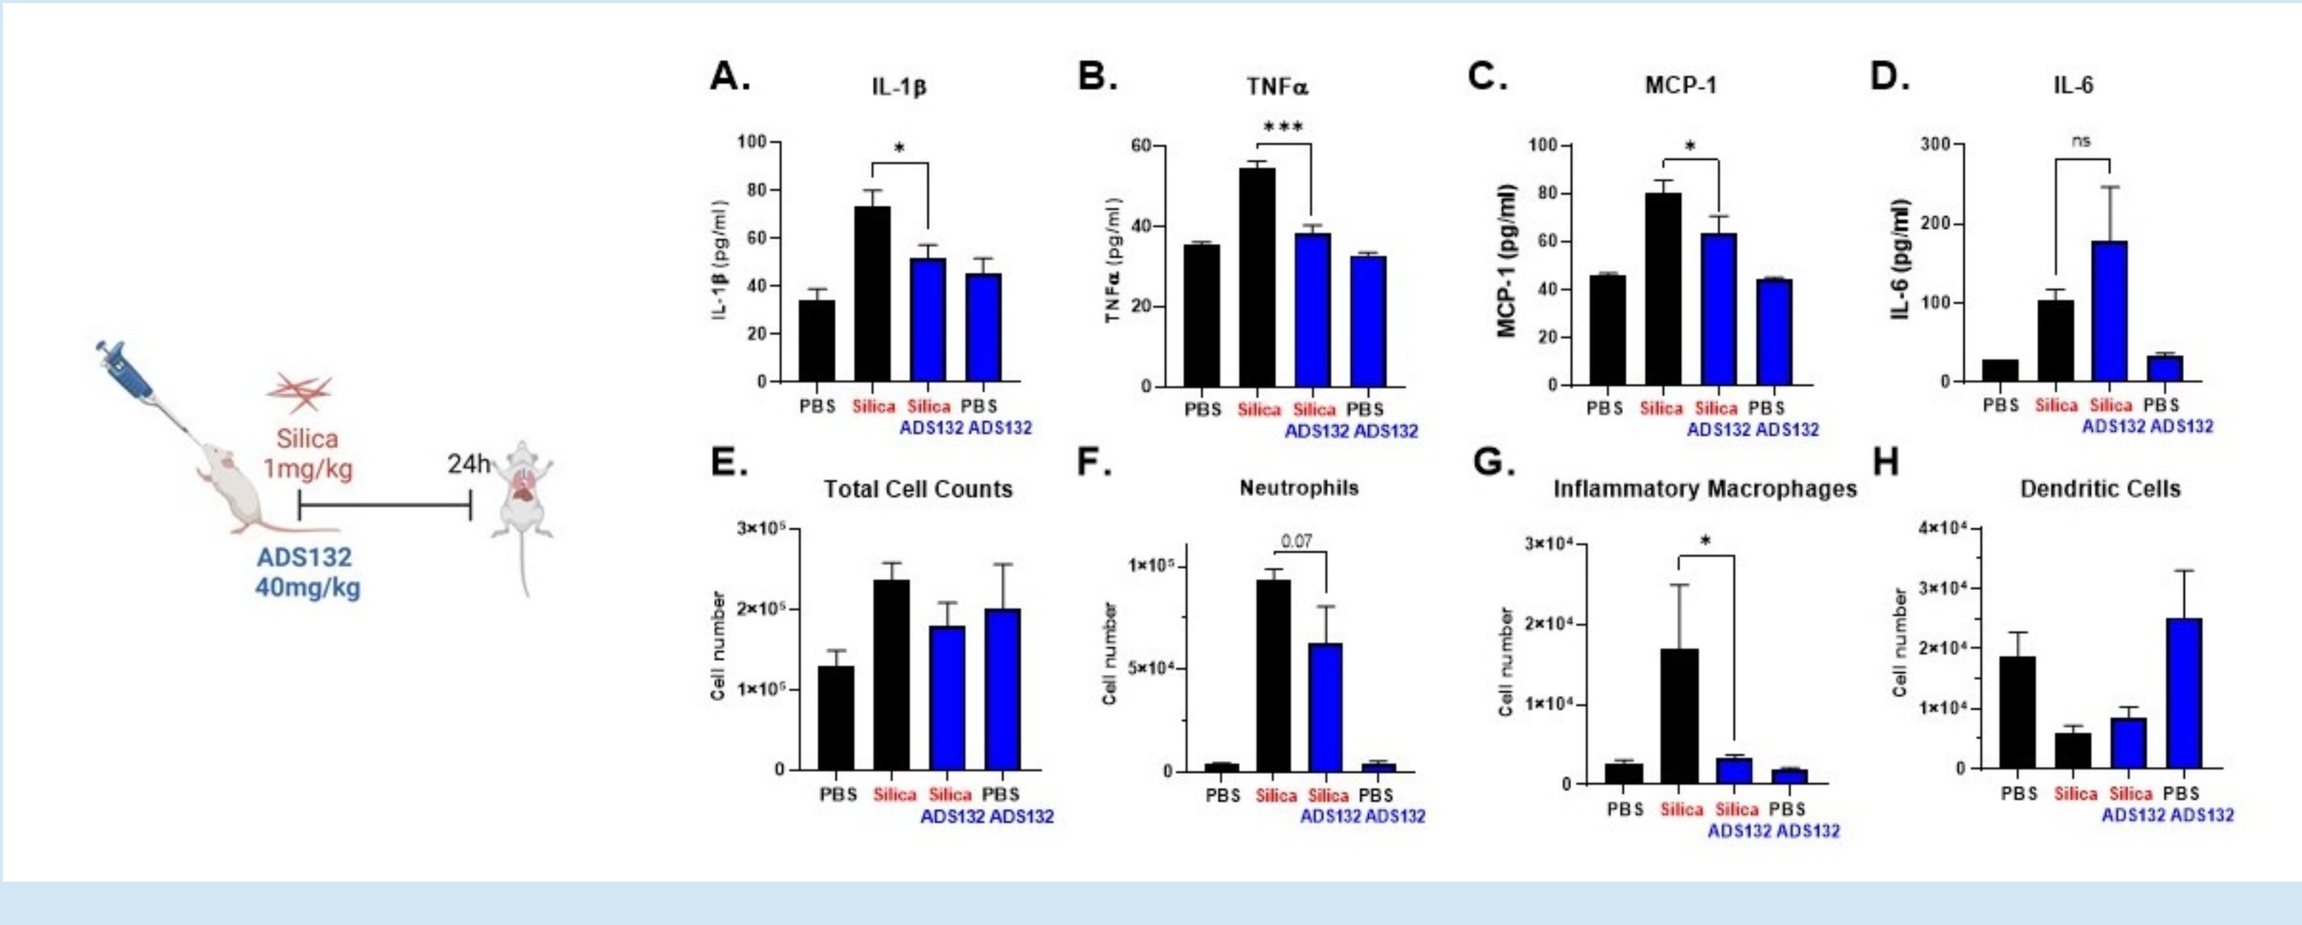
Silica 1mg/kg
24h
ADS132 40mg/kg
A. IL-1β
B. TNFα
C. MCP-1
D. IL-6
E. Total Cell Counts
F. Neutrophils
G. Inflammatory Macrophages
H Dendritic Cells

ADS132 reduces pulmonary inflammation in an NLRP3-dependent occupational lung disease model

Wild type C57Bl/6 mice (n=6 per treatment group) received either PBS alone, silica (1mg), silica (1mg) mixed with ADS132 (40mg/kg) in PBS, or ADS0132 via intranasal inoculation. Twenty-four hours later, mixed were killed and bronchial lavage fluid harvested. Proinflammatory cytokines. Data are presented are the means ± SEM. *p < 0.05, ***p < 0.001, n.s.- not significant. One-way ANOVA, Dunnett's multiple comparison.

**ADS132 is a PEGylated version of ADS032 that retains similar biological activity.*

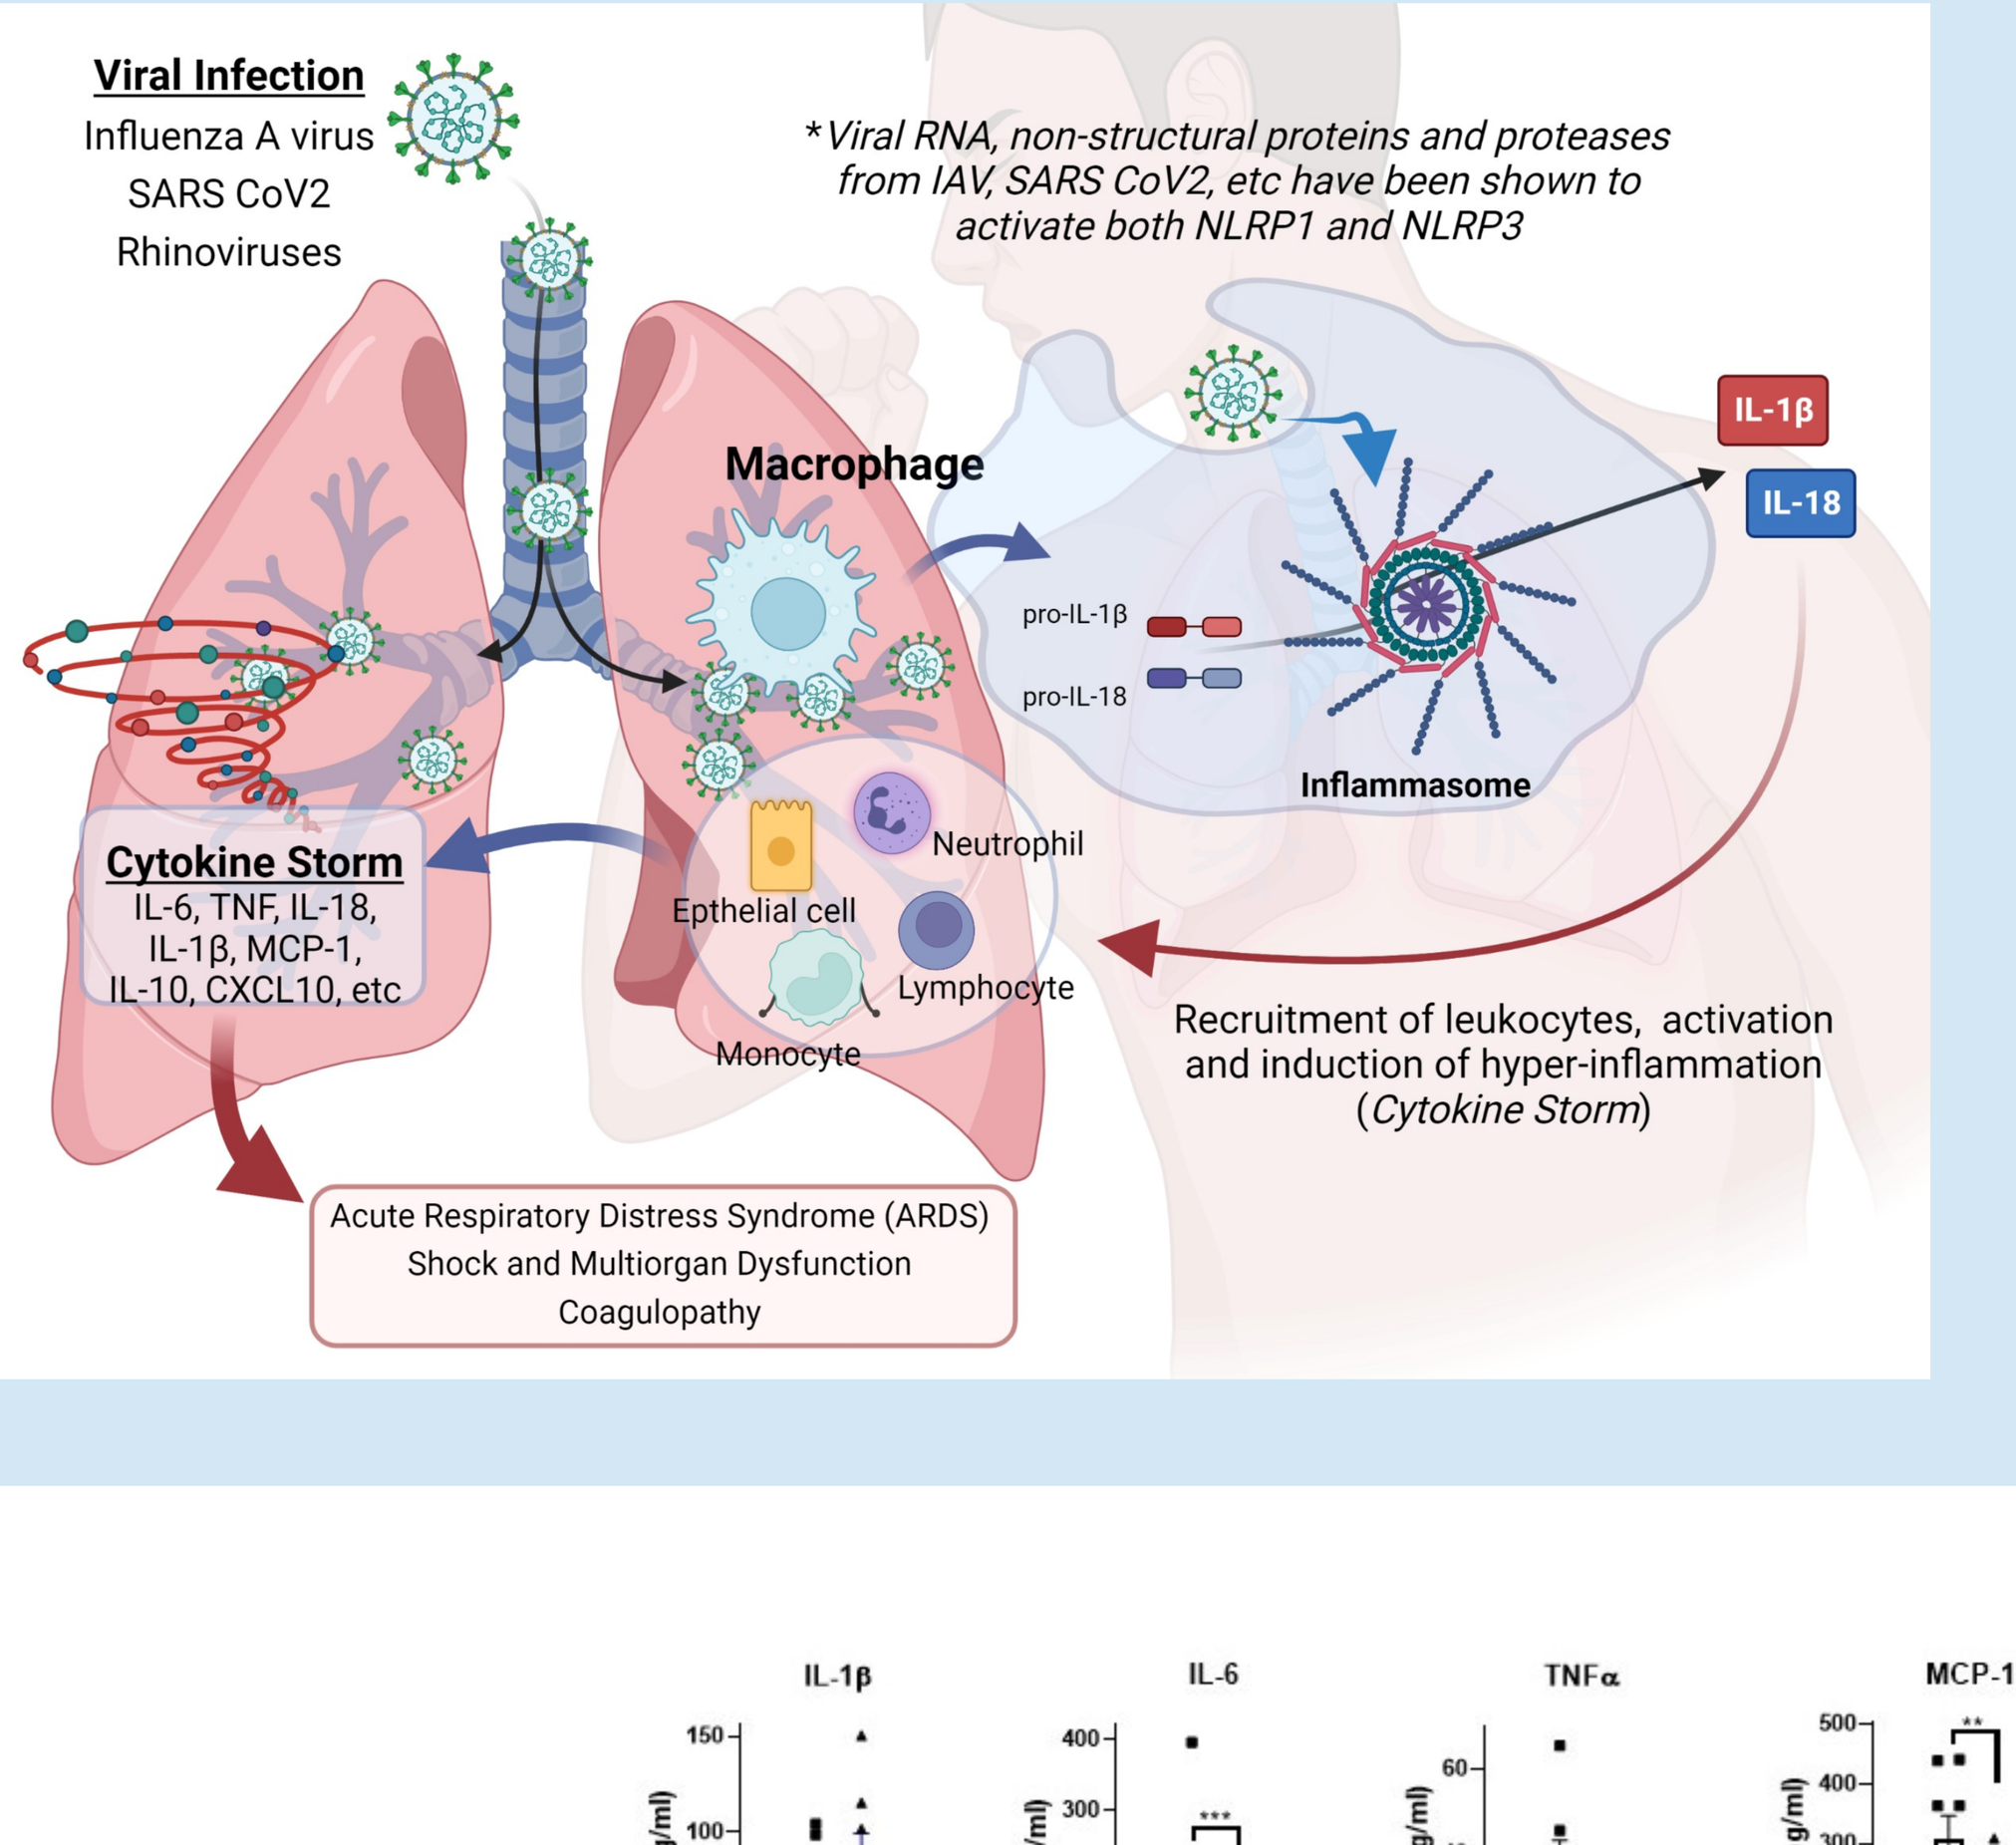
Viral Infection
Influenza A virus
SARS CoV2
Rhinoviruses
*Viral RNA, non-structural proteins and proteases from IAV, SARS CoV2, etc have been shown to activate both NLRP1 and NLRP3
Macrophage
pro-IL-1β
pro-IL-18
IL-1β
IL-18
Inflammasome
Cytokine Storm
IL-6, TNF, IL-18, IL-1β, MCP-1, IL-10, CXCL10, etc
Epthelial cell
Neutrophil
Lymphocyte
Monocyte
Recruitment of leukocytes, activation and induction of hyper-inflammation (Cytokine Storm)
Acute Respiratory Distress Syndrome (ARDS)
Shock and Multiorgan Dysfunction
Coagulopathy

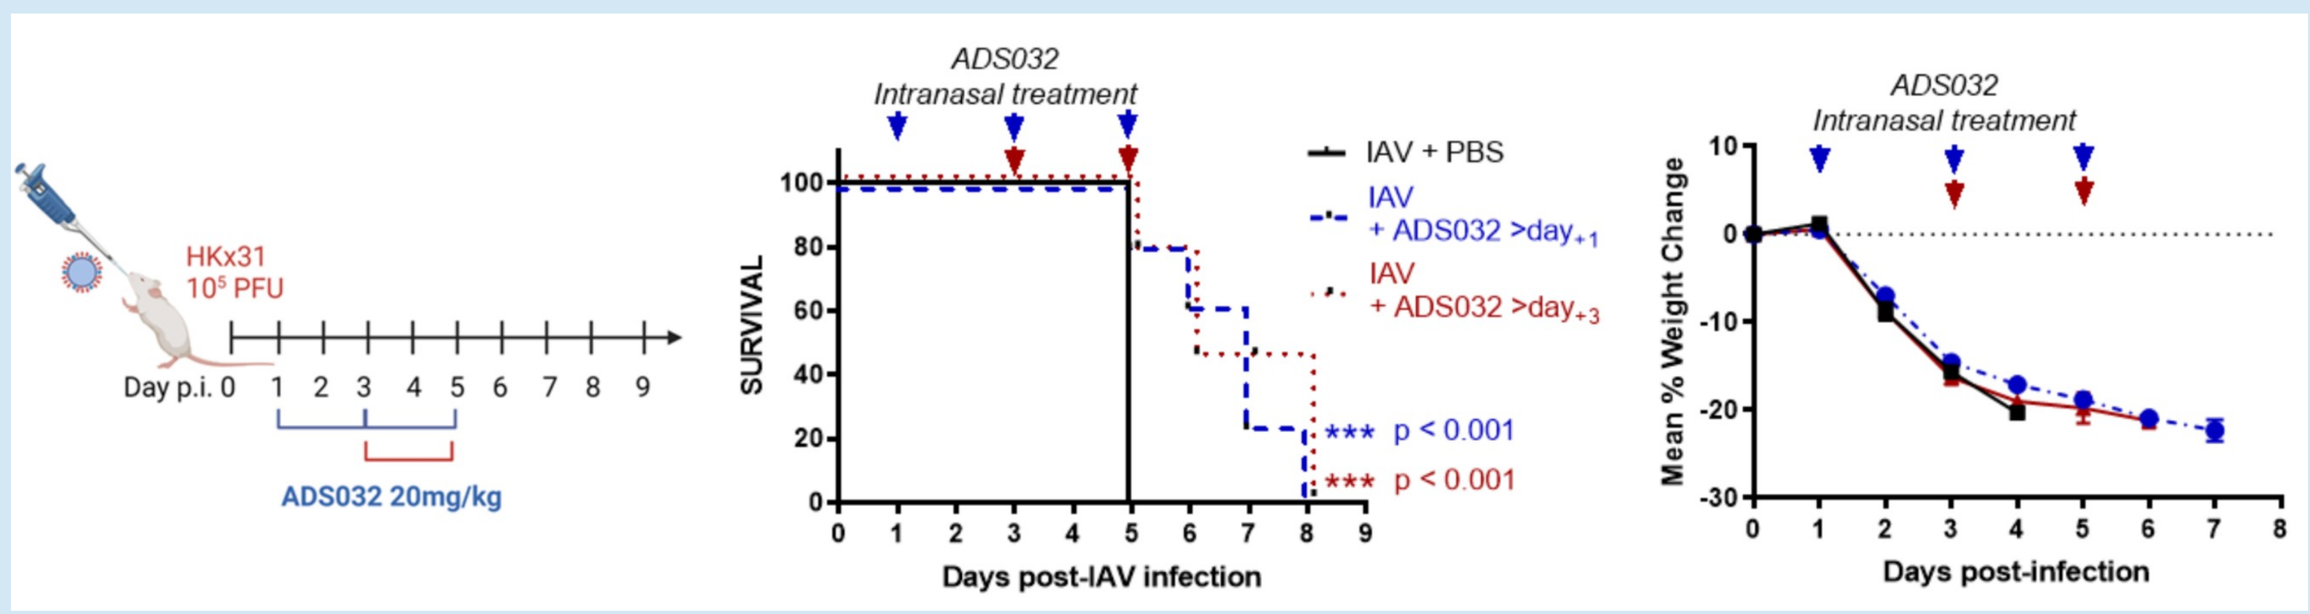
HKx31
10^5 PFU
Day p.i. 0
ADS032 20mg/kg
ADS032 Intranasal treatment
IAV + PBS
IAV + ADS032 >day+1
IAV + ADS032 >day+3
SURVIVAL
*** p < 0.001
*** p < 0.001
Days post-IAV infection
Mean % Weight Change
Days post-infection

ADS032 administered at any time protects mice from pathogenic influenza A virus (IAV) infection.

Groups of wild-type male and female C57Bl/6 mice (IAV/PBS n=8; IAV/ADS032 > Day+1 n=14; IAV/ADS032 > Day +3 n=8) were intranasally challenged with HKx31 (10^5 PFU) IAV.
Mice were treated intranasally with PBS (IAV) or ADS032 (20mg/kg) from either day 1 or day 3 posts-infection and every 48 hr thereafter until day 5 post-infection. Mice were weighed daily, and results were expressed as percentage of weight lost as Day 0, the mean ± SEM.
Survival curves are shown. ***P < 0.001. Mantel-Cox log-rank test.

NOTE: ***Timing of MCC950 treatment profoundly affects disease outcome.*** <u>While ADS032-treated mice are protected when treated at any time during infection</u>; we have previously published that MCC950-treated mice are hypersusceptible to IAV infection when treated from Day 1 post-infection, but protected when treated from day 3 post-infection (*Tate et al, 2017, Sci Reports, 2: 27912*)

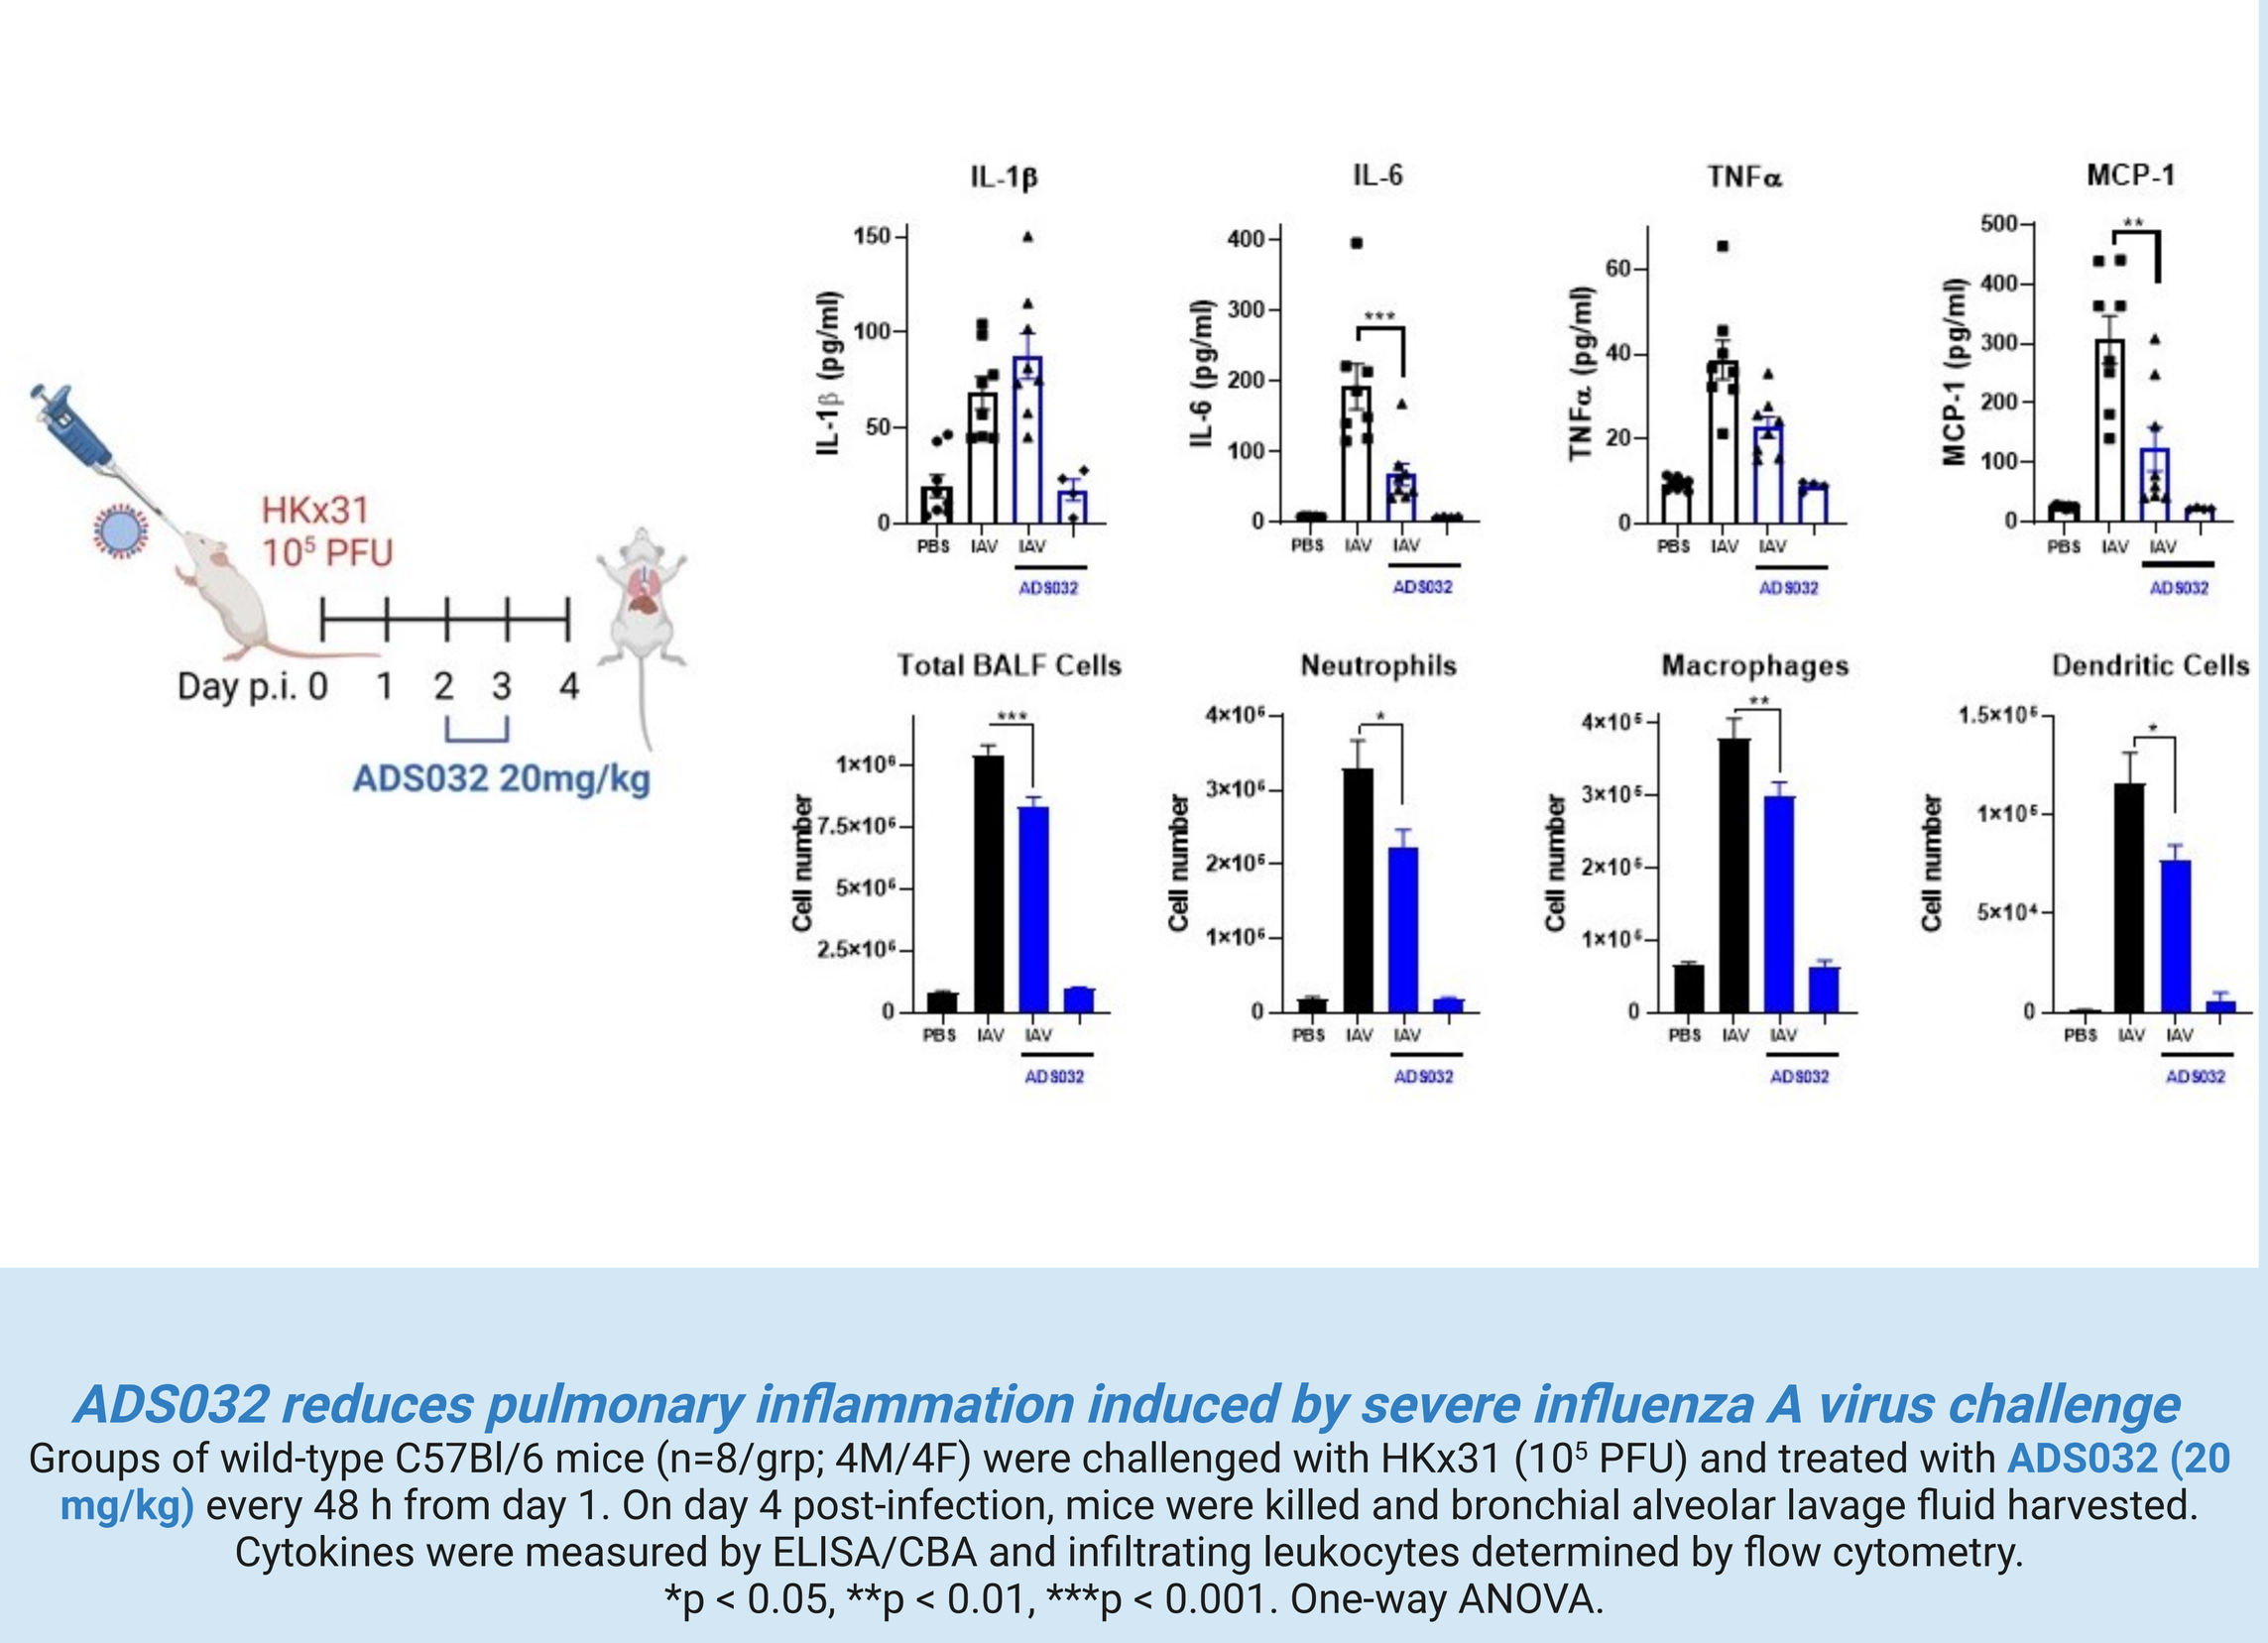
HKx31
10^5 PFU
Day p.i. 0 1 2 3 4
ADS032 20mg/kg
IL-1β
IL-6
TNFα
MCP-1
Total BALF Cells
Neutrophils
Macrophages
Dendritic Cells

ADS032 reduces pulmonary inflammation induced by severe influenza A virus challenge

Groups of wild-type C57Bl/6 mice (n=8/grp; 4M/4F) were challenged with HKx31 (10^5 PFU) and treated with ADS032 (20 mg/kg) every 48 h from day 1. On day 4 post-infection, mice were killed and bronchial alveolar lavage fluid harvested. Cytokines were measured by ELISA/CBA and infiltrating leukocytes determined by flow cytometry.
*p < 0.05, **p < 0.01, ***p < 0.001. One-way ANOVA.

CONCLUSION

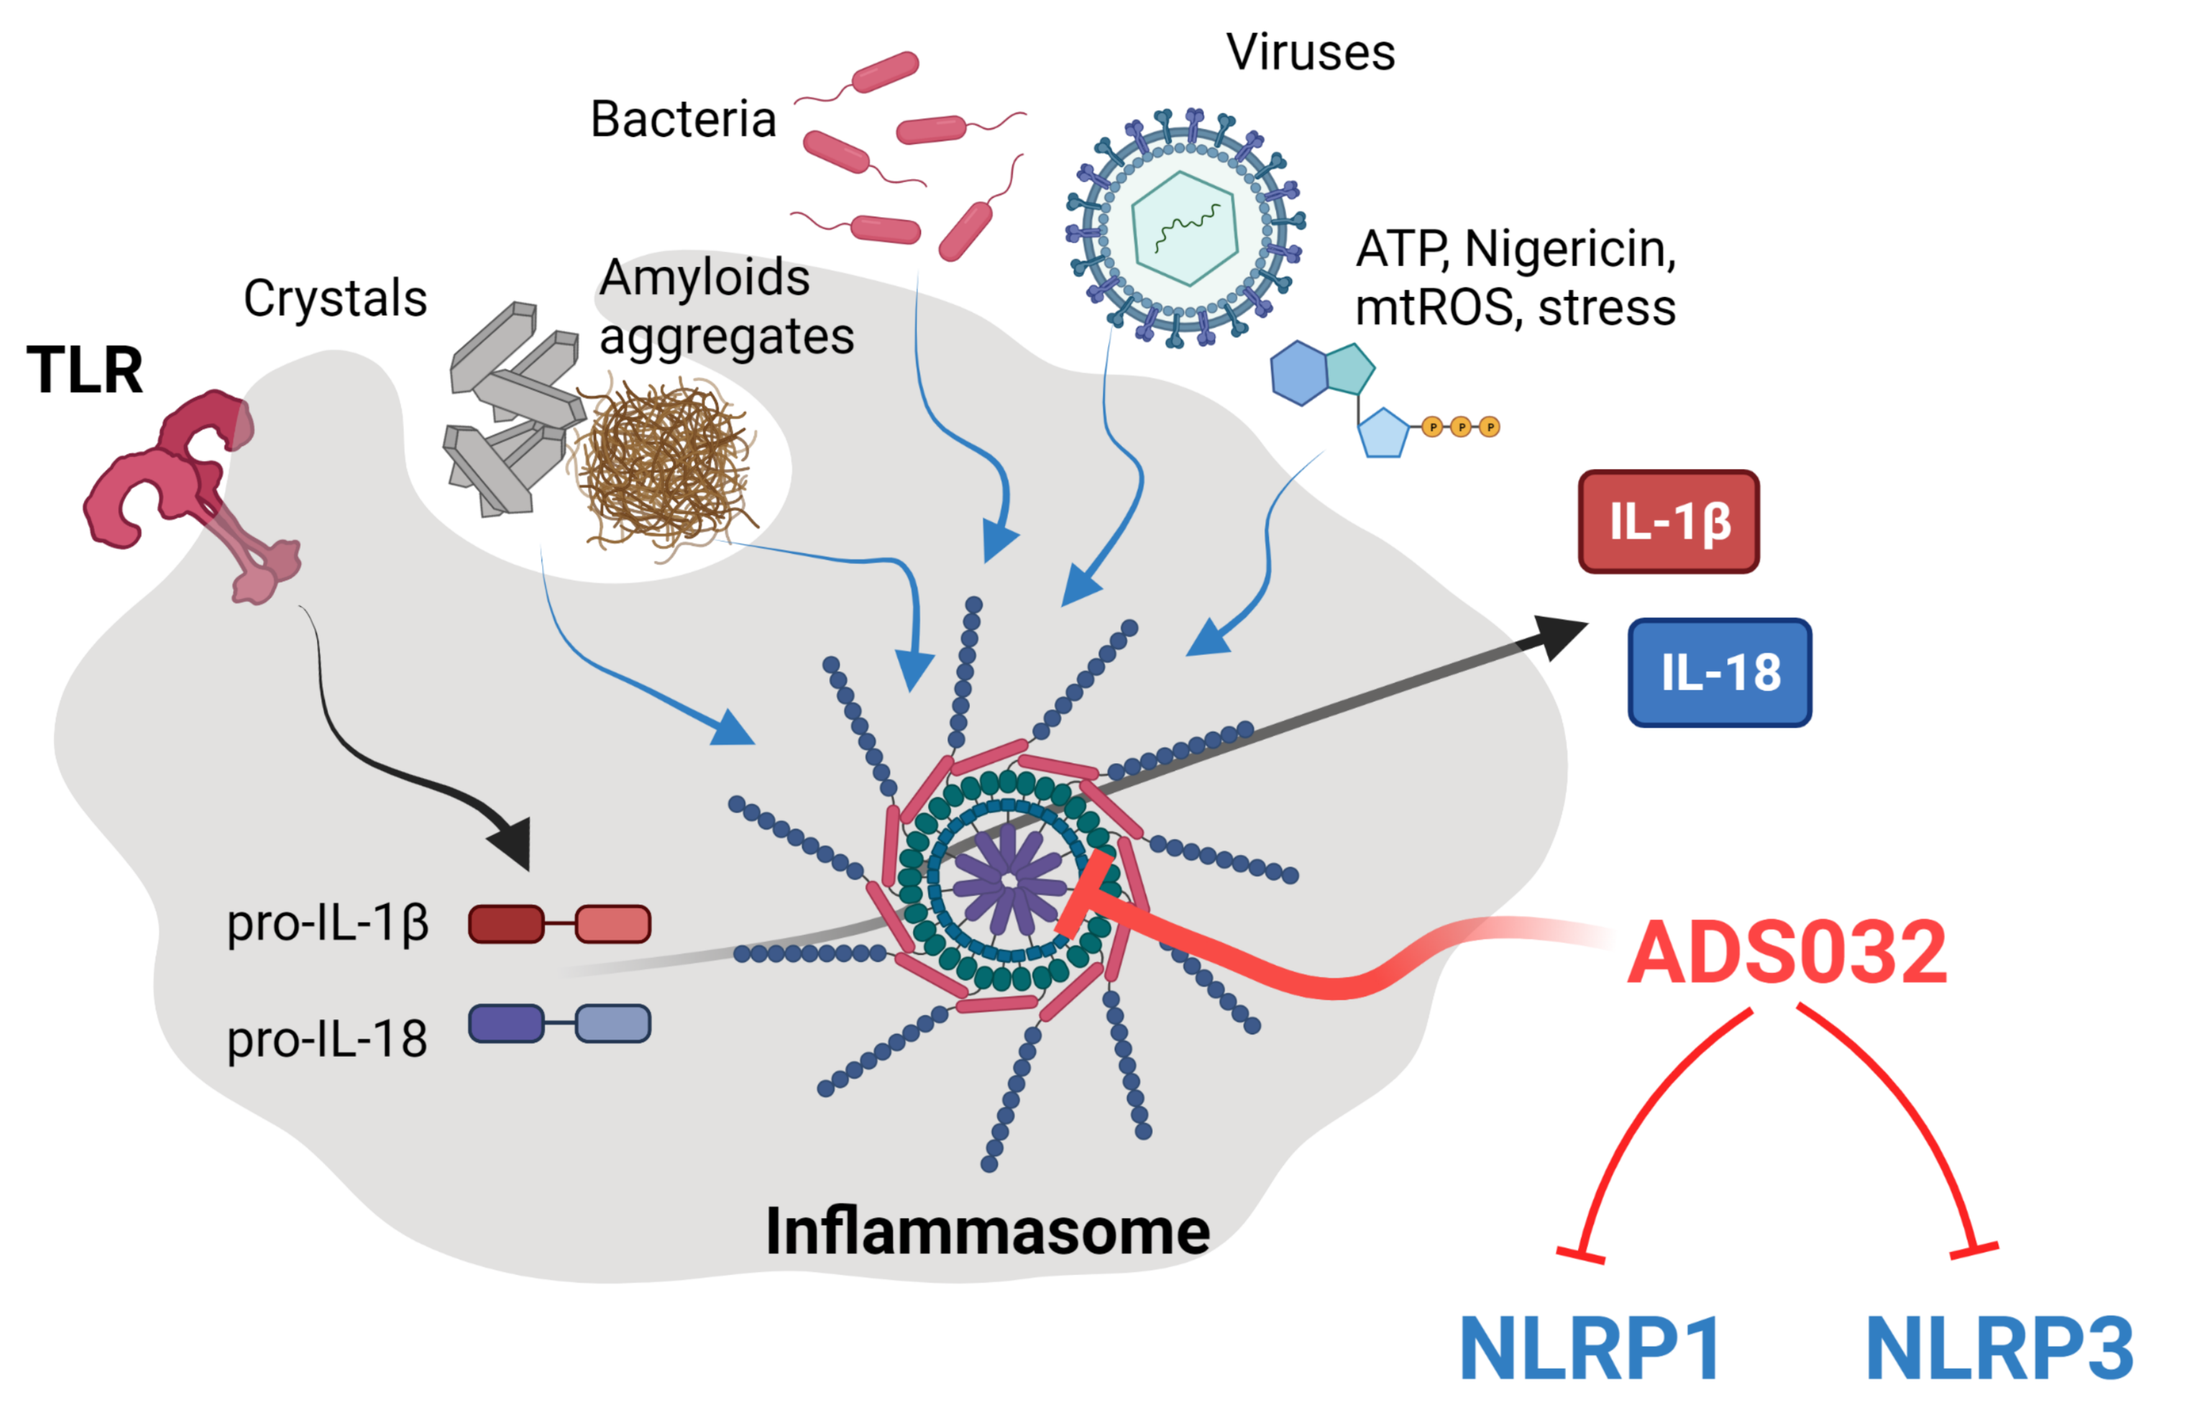
Viruses
Bacteria
ATP, Nigericin, mtROS, stress
Crystals
Amyloids aggregates
TLR
IL-1β
IL-18
pro-IL-1β
pro-IL-18
ADS032
Inflammasome
NLRP1
NLRP3

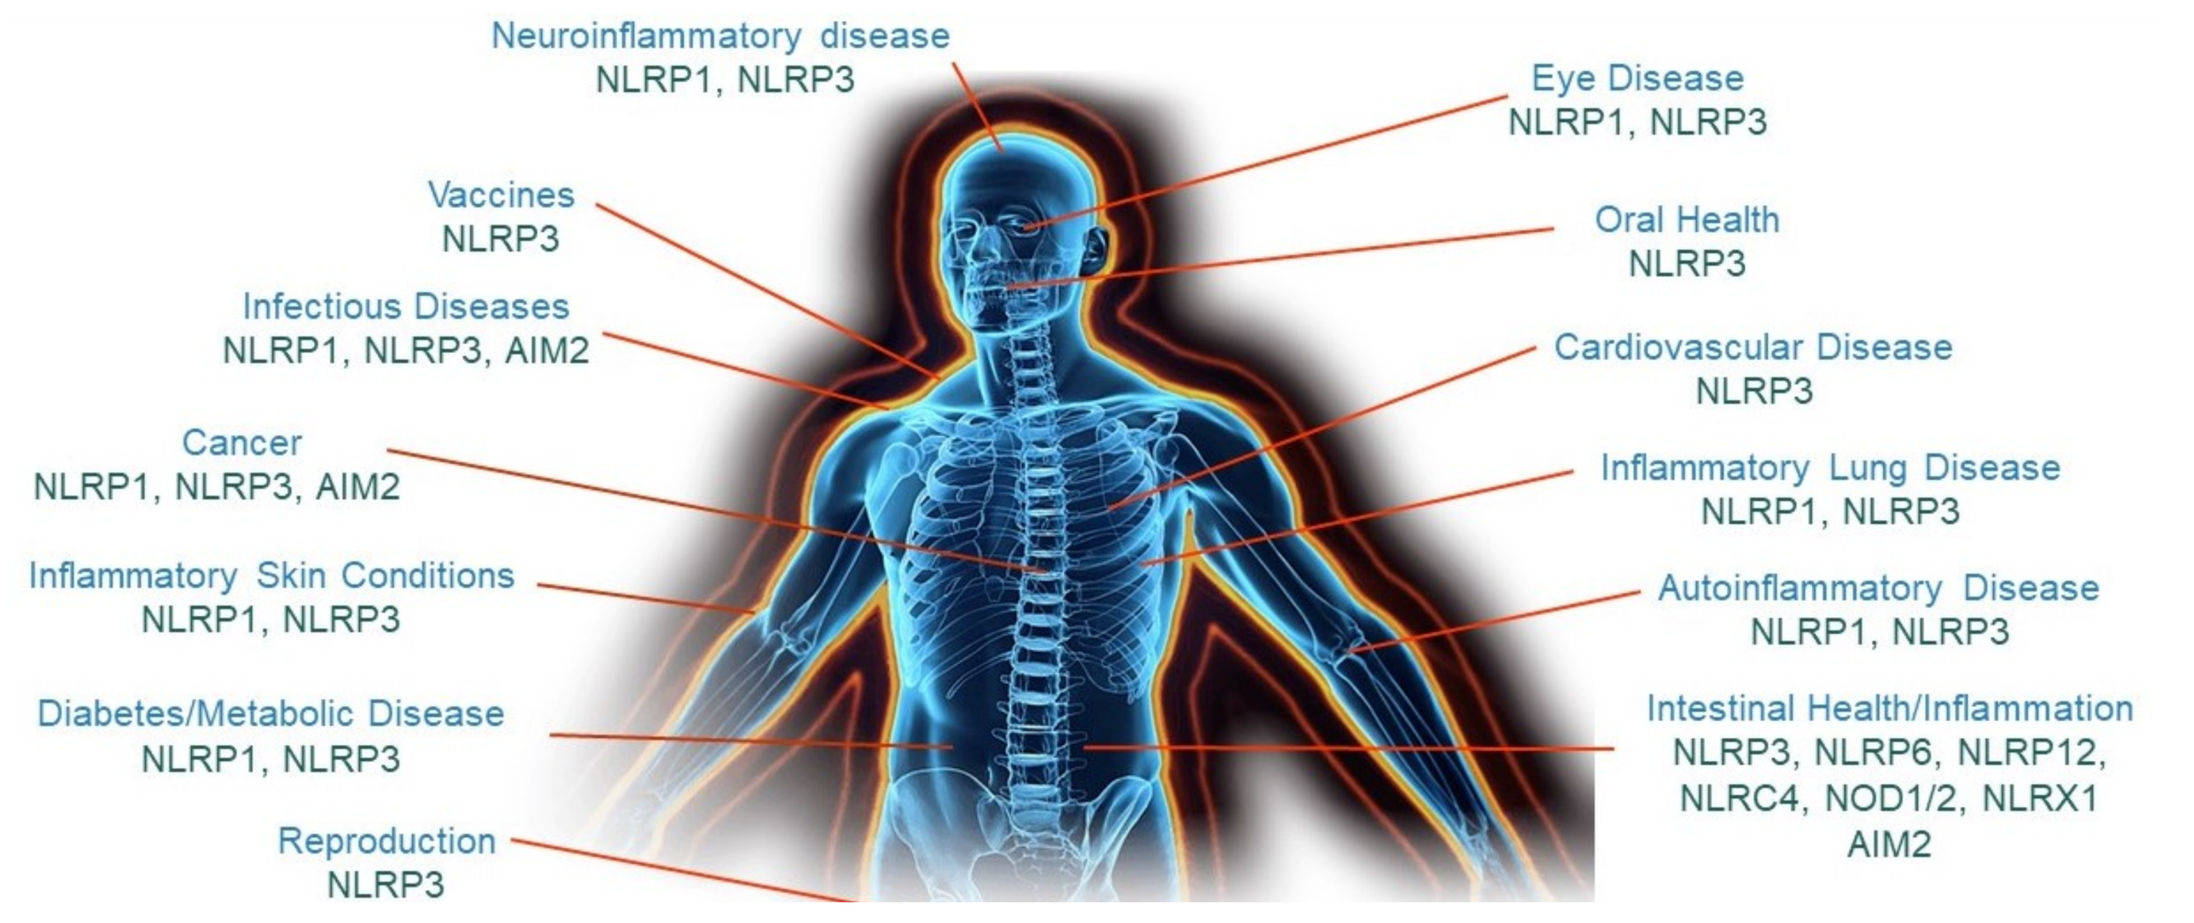
Neuroinflammatory disease NLRP1, NLRP3
Eye Disease NLRP1, NLRP3
Vaccines NLRP3
Oral Health NLRP3
Infectious Diseases NLRP1, NLRP3, AIM2
Cardiovascular Disease NLRP3
Cancer NLRP1, NLRP3, AIM2
Inflammatory Lung Disease NLRP1, NLRP3
Inflammatory Skin Conditions NLRP1, NLRP3
Autoinflammatory Disease NLRP1, NLRP3
Diabetes/Metabolic Disease NLRP1, NLRP3
Intestinal Health/Inflammation NLRP3, NLRP6, NLRP12, NLRC4, NOD1/2, NLRX1 AIM2
Reproduction NLRP3

INFLAMMASOME-ASSOCIATED DISEASE

What is known

- Inflammasome-induced inflammation is associated with a wide range of acute and chronic diseases
- There is intense interest in developing inhibitors and therapeutics to reduce inflammasome-mediated inflammation in disease

What this study adds

- It identifies the first described dual NLRP1 and NLRP3 inhibitor
- ADS032 is an effective NLRP1 and NLRP3 antagonist in human macrophages and epithelial cells
- Treatment of mice with ADS032 reduces LPS-induced systemic inflammation, and silica- and IAV-mediated pulmonary disease

Clinical significance

- ADS032 may provide clinical benefit in addressing inflammasome-associated viral and occupational inflammatory disease
- This study identifies ADS032 as a potential therapeutic to treat multiple NLRP1- and NLRP3-associated inflammatory diseases
- ADS032 is the first human NLRP1 antagonist allowing examination of NLRP1 in human disease

Created with BioRender Poster Builder